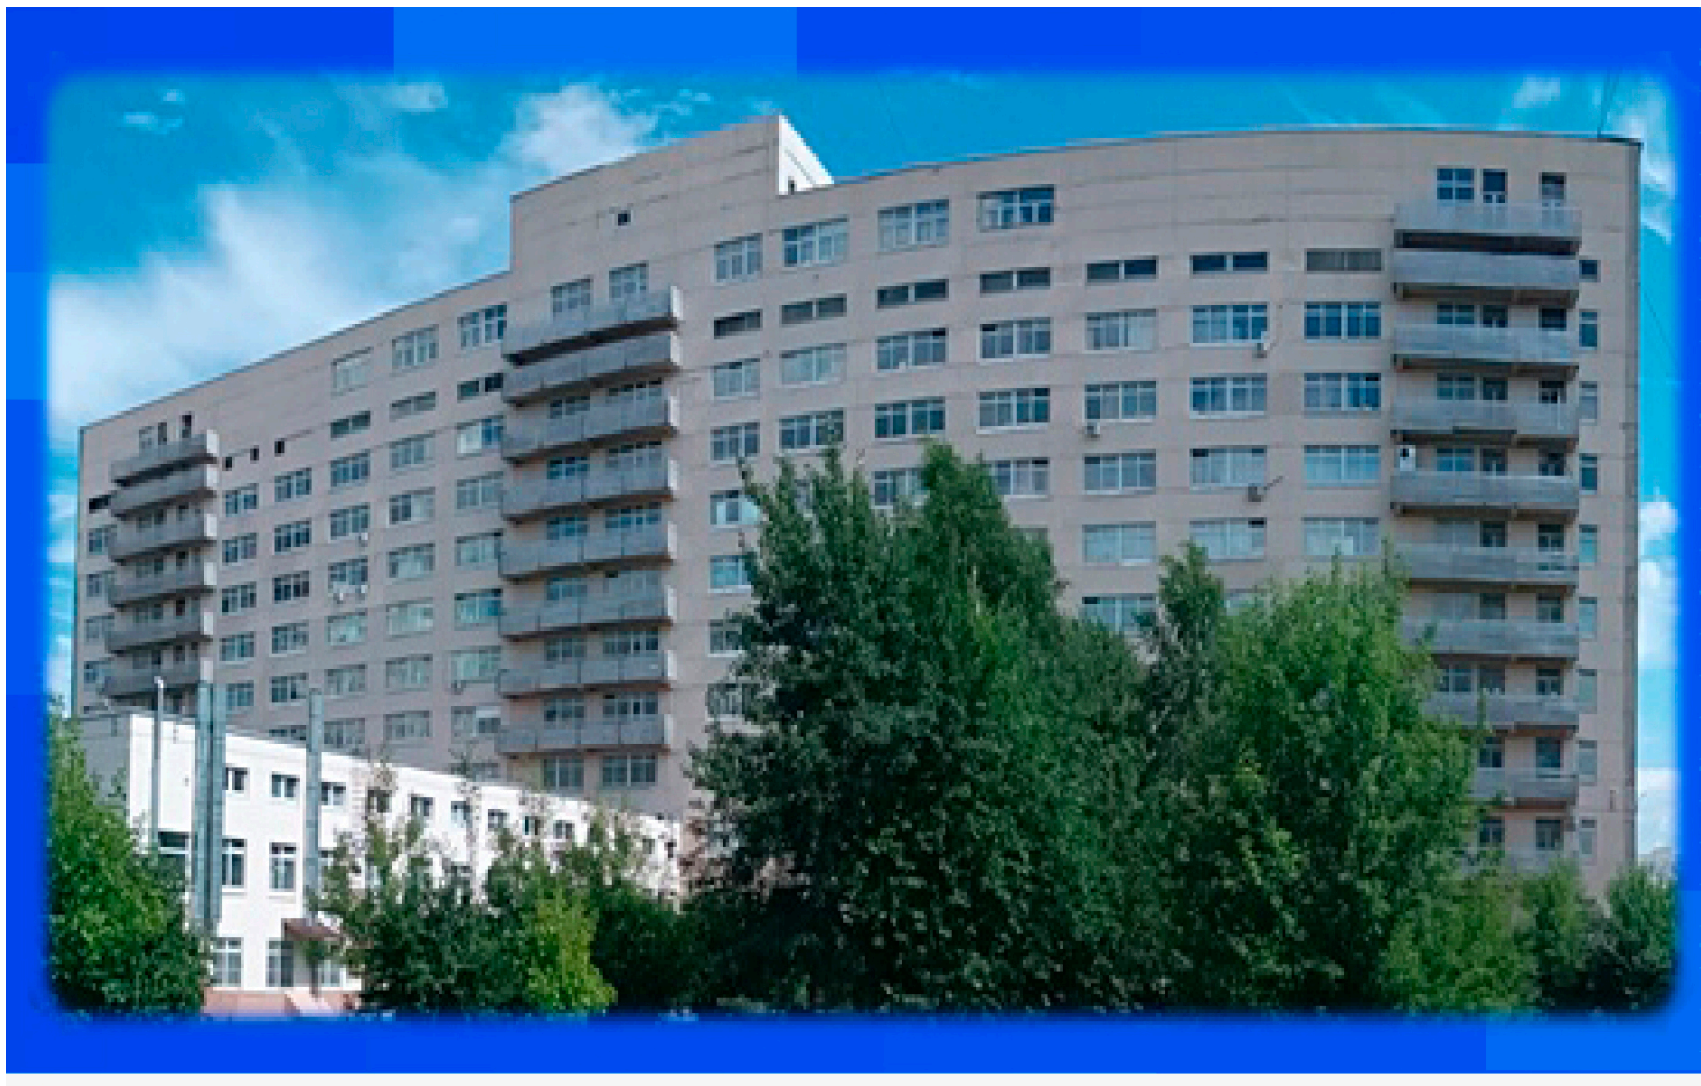

EUROPEAN E-CONGRESS OF RHEUMATOLOGY 2020, From 3 June

№ SAT0427

Ccomparative effectiveness of tofacitinib and adalimumab in psoriatic arthritis patients in real clinical practice.

Loginova E.Yu., Korotaeva T.V., Gubar E.E., Korsakova Y.L., Glukhova S.I.,

on behalf of the RU-PsART study group

Background.

Tofacitinib (TF) is an oral Janus kinase inhibitor approved for the PsA patients (pts) treatment. It has demonstrated comparable efficacy to ADA in RCT across PsA clinical symptoms. [1,2] There is currently no data on comparative effectiveness of TF and ADA in a real clinical practice. The **R**ussian **PsA** **R**egis**T**ry (RU-PsART) collected data from 43 rheumatology centers in the Russian Federation.

Objectives.

To investigate 6 months (mo) treatment responses to TF or ADA in pts with active PsA in a real clinical practice.

Methods.

77 (M/F=43/34) PsA pts according to CASPAR criteria from the RU-PsART cohort were divided into two groups according to treatment. 41pts (M/F=24 (58.5%)/17 (41.5%), mean age 42.4±10,3 years (yrs), median (Me) PsA duration 72 [35;120] mo were treated with TF 5 mg twice daily. 36 pts (M/F=19 (52.8%)/17 (47.2%), mean age 44±11.5 yrs, Me PsA duration 59 [22;102] mo were treated with ADA 40 mg/2 weeks subcutaneous as first-line bDMARDs. In TF/ADA groups 89%/52.8% pts were taking combination therapy with Methotrexate (MTX) accordingly. At baseline (BL) and 6 mo of therapy the PsA activity by DAPSA, BASDAI, the number of pts (NPts) with dactylitis, enthesitis by LEI, BSA, HAQ were evaluated in both groups. At 6 mo the NPts achieved remission DAPSA (≤4)/Low Activity (LA)≤14) and Minimal disease activity (MDA, 5 of 7) were calculated in both groups. M±SD, Me [Q25; Q75], Min-Max, %, t-test, Pierson-χ^2, Manna-Whitney, Wilcoxon tests were performed. All $p<0.05$, were considered to indicate statistical significance.

Results.

At BL TF pts had higher PsA activity by DAPSA compare to ADA group: 44.2[37.8; 55.3]/35.8 [21.1; 52] accordingly (p=0.03). At 6 mo of treatment DAPSA significantly decreased in TF/ADA groups - to 11 [4,3; 17,3]/9,1[6; 19,6] (p<0,001) accordingly (Fig.1).

At 6 mo of therapy there were no significant differences between groups in achieving MDA, DAPSA LA/Remission (table 1).

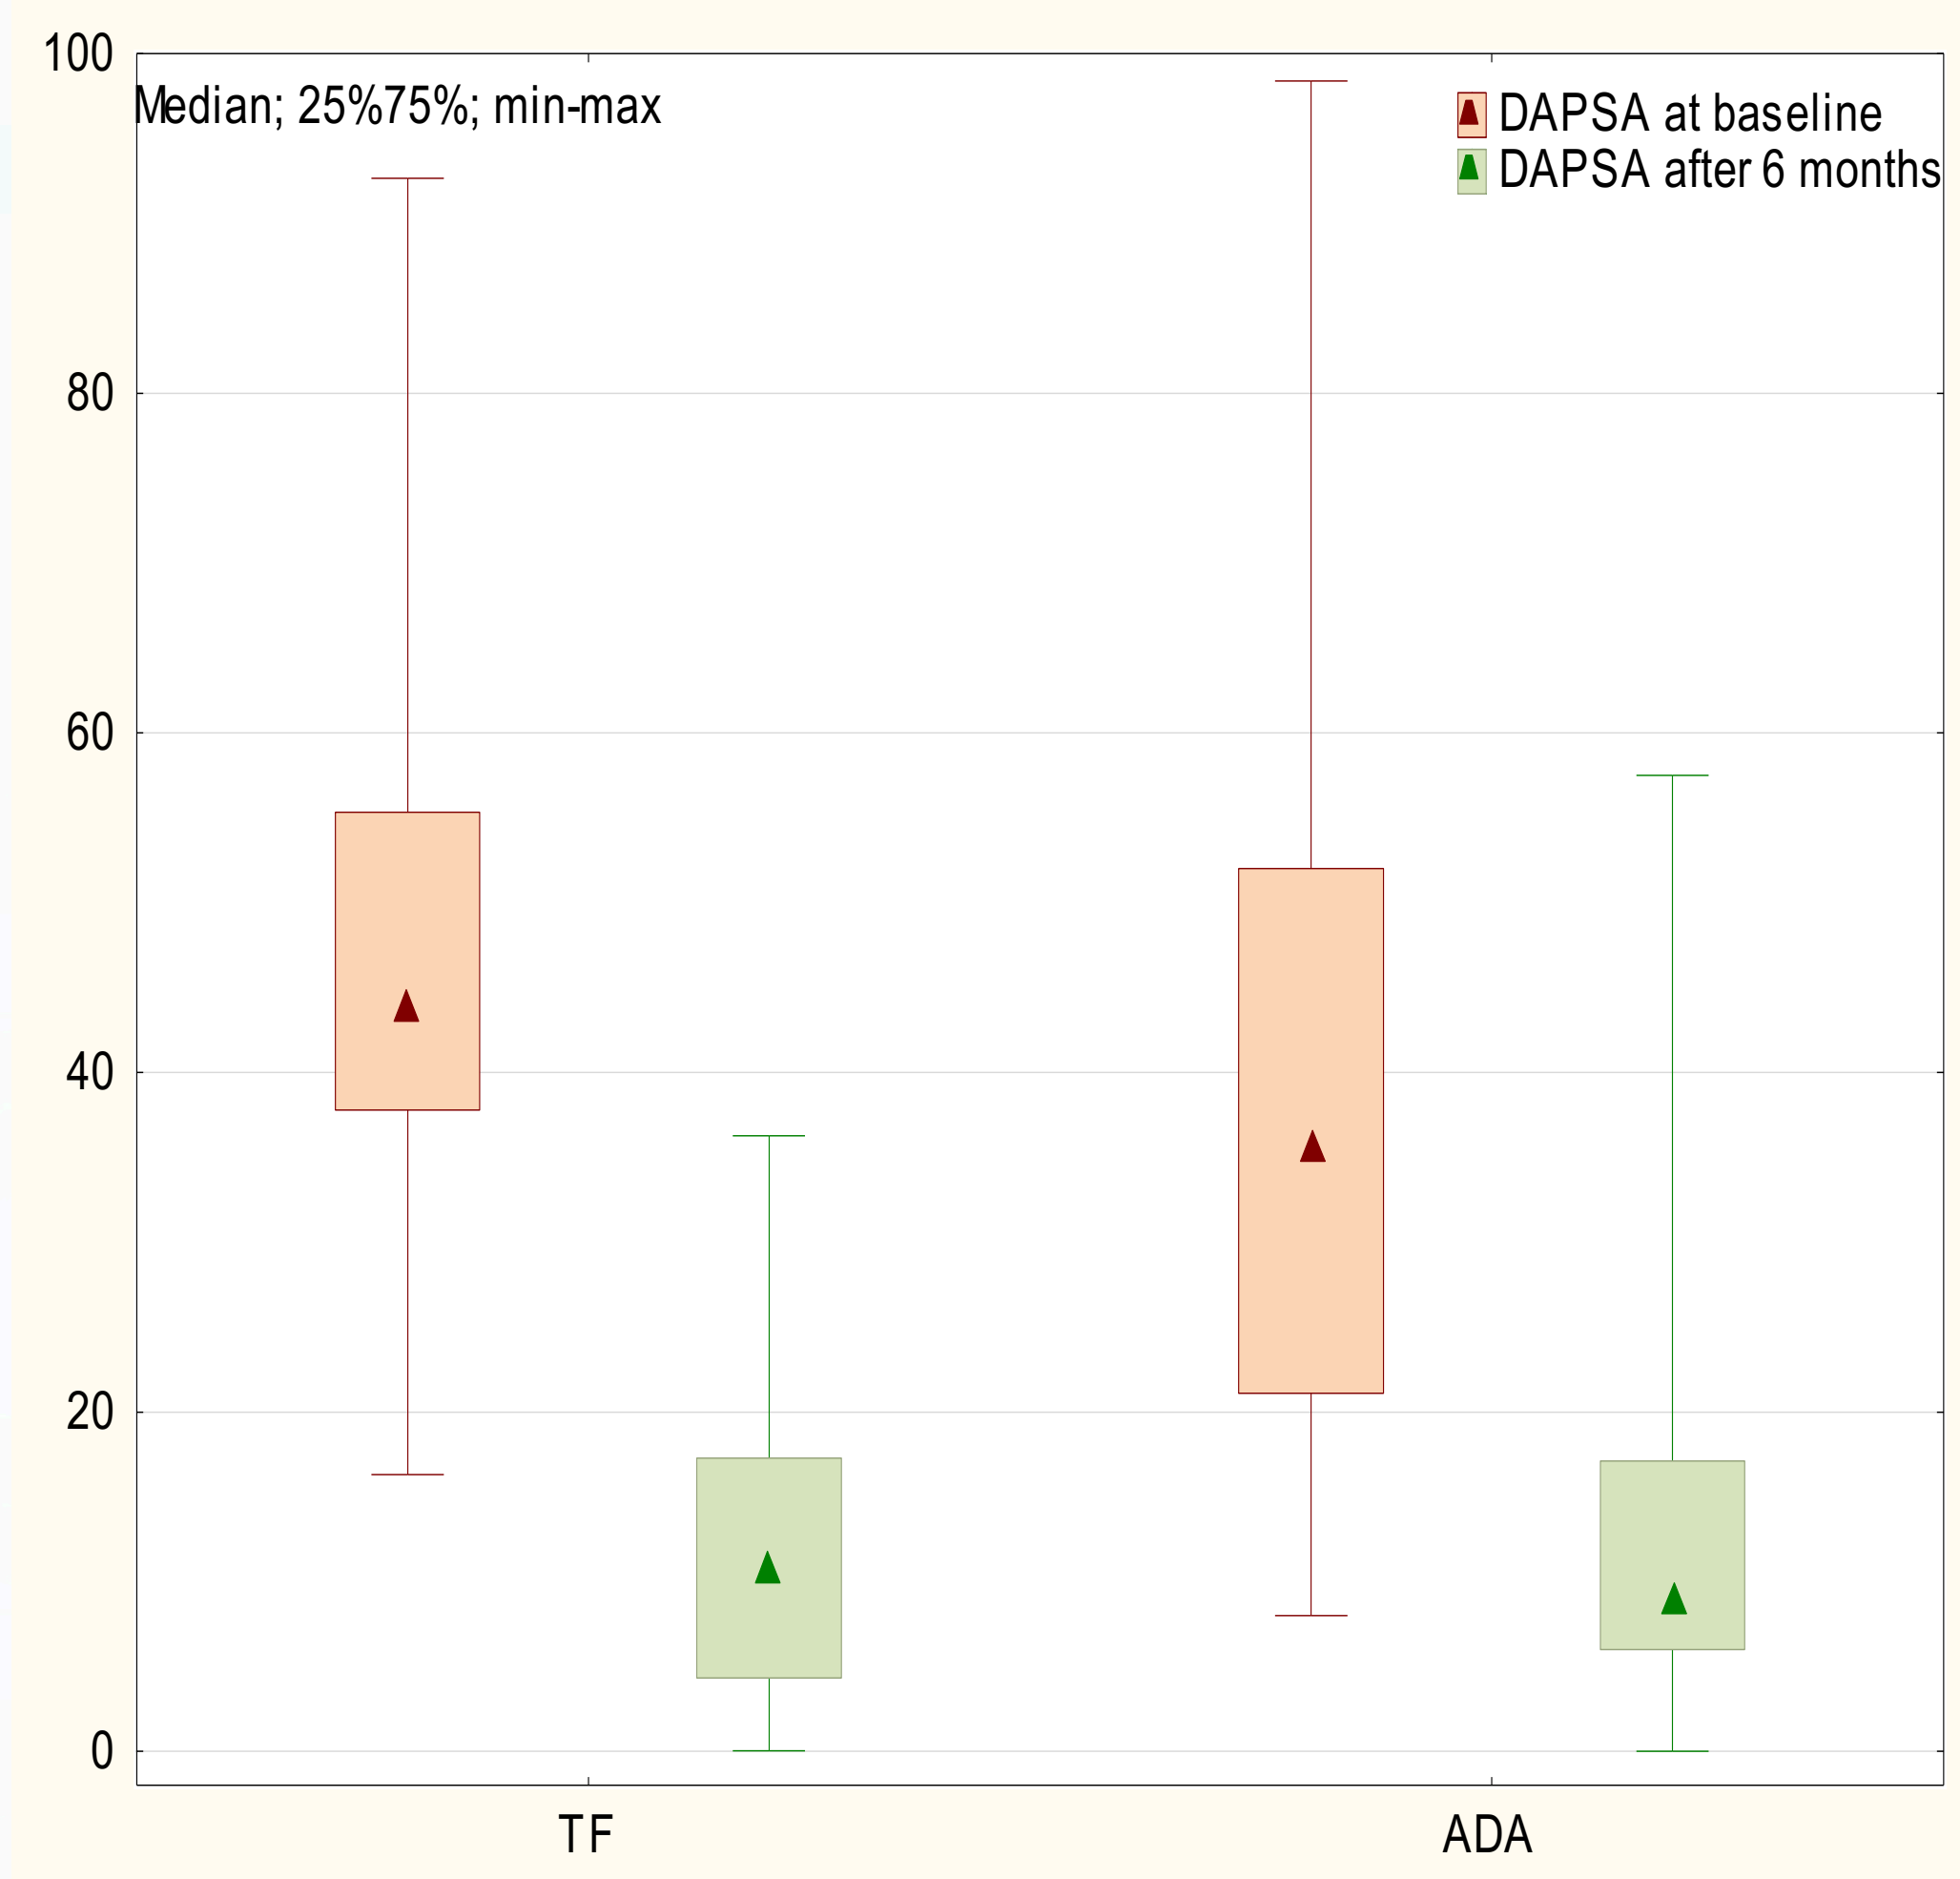
100
80
60
40
20
0
Median; 25%75%; min-max
DAPSA at baseline
DAPSA after 6 months
TF
ADA

Fig.1. DAPSA in TF/ADA at 6 months

	TF		ADA			
Study	Events	Subjects	Events	Subjects	OR (95% CI)	P
DAPSA remission abs, % achieved	11 (26,8%)	41	6 (20,8%)	29	1,406 (0,453-4,366)	0.556
DAPSA LA abs, % achieved	15 (36,6%)	41	13 (44,8%)	29	0,710 (0,269-1,872)	0.489
MDA abs, % achieved	16 (40%)	40	9 (30%)	30	1,556 (0,569-4,249)	0.389

Table1. MDA, DAPSA LA/Remission in TF/ADA at 6 months

At 6 mo of treatment in TF-treated group the NPts with dactylitis significantly decreased from 22 (53,7%) to 5 (13,2%) pts (p<0.001) and in ADA-treated group no significantly differences were found in NPts with dactylitis, despite a quantitative decrease: from 13 (36,1%) to 6 (20%) pts (p=0.15). The NPts with enthesitis significantly decreased in both TF/ADA groups: from 27 (69,2%) to 12 (30,8%) (p<0,002) and from 16 (72,7%) to 5 (38,5%) (p<0.046) pts accordingly. In TF/ADA-treated pts BASDAI improved from 6 [4.2;7] to 1.4 [0.6; 3.2] (p<0.001)/from 4.4 [1.9;5.8] to 3 [0.8;4.5] accordingly (p<0.046); HAQ significantly decreased from 1 [0.625;1.5] to 0,5 [0; 0.875] and from 0.875 [0.5;1.38] to 0.5 [0; 0.875] accordingly. The NPts with BSA>3% significantly reduced from 20 (51,3%) to 8 (26,7%) pts (p<0,002) in TF group only compared to ADA.

Conclusion.

In active PsA TF and ADA provide comparable effectiveness across all clinical domains and attainment of MDA/LA/Remission by DAPSA in a real clinical practice.

References.

1. Mease P, Hall S, FitzGerald O, et al. N Engl J Med 2017;377:1537–50. 13.
2. Gladman D, Rigby W, Azevedo VF, et al. N Engl J Med 2017;377:1525–36.

V. A. Nasonova Research Institute of Rheumatology, Moscow, Russian Federation, +7 (495)109-29-10, sokrat@irramn.ru, https://rheumatolog.su